Phase 1 Study to Evaluate the Safety, Tolerability and Preliminary Efficacy of Rivoceranib Plus Paclitaxel in Advanced Gastric or Gastroesophageal Junction Cancer

Presented at ESMO 2022
1237P

Min-Hee Ryu,[1] Jae Hong Kim,[2] Yoon-Koo Kang[1]

[1]Asan Medical Center, University of Ulsan College of Medicine, Seoul, Republic of Korea; [2]Elevar Therapeutics, Salt Lake City, UT, USA

INTRODUCTION

- Rivoceranib is a novel oral tyrosine kinase inhibitor that potently and selectively inhibits vascular endothelial growth factor receptor 2 (VEGFR2)[1,2]
- VEGFR2 demonstrates greater kinase activity than VEGFR1/3 and represents the primary receptor for regulation of mitogenic signaling, angiogenesis, and vascular permeability[3]
- Rivoceranib is approved in China as apatinib for later-line treatment of recurrent or metastatic gastric and hepatocellular carcinoma[4]
- Rivoceranib enhances the chemosensitivity of gastric cancer to paclitaxel and 5-fluorouracil *in vitro* and *in vivo*[5]
- We conducted a phase 1/2a study to evaluate the safety, tolerability, and preliminary efficacy of rivoceranib in combination with paclitaxel in advanced gastric or gastroesophageal junction (GEJ) cancer
- Herein we report the phase 1 results

OBJECTIVE

- The primary objective of the phase 1 portion was to determine the recommended phase 2 dose (RP2D) of rivoceranib in combination with paclitaxel

METHODS

- Open-label, single-arm, dose-escalation (standard 3+3) phase 1 study (NCT03707028) (**Figure 1**)
- Patients received rivoceranib in combination with paclitaxel in 4-week cycles
- The starting dose was rivoceranib 400 mg orally once daily in combination with intravenous (IV) paclitaxel 80 mg/m² administered on days 1, 8, and 15 of the 28-day cycle. Premedication according to standard clinical practice at the study center was required prior to the first 2 weekly doses and stopped if no infusion reaction was observed
- Planned dose level 1, 2, and 3 are paclitaxel 80 mg/m² in combination with rivoceranib 400 mg, 500 mg, and 600 mg, respectively. In case dose level 1 is not tolerable, paclitaxel could be reduced to 60 mg/m² (dose level -1) or 50 mg/m² (dose level -2)
- Tumors were assessed using Response Evaluation Criteria in Solid Tumors (RECIST) v1.1 every 8 weeks
- Adverse events were evaluated according to Common Terminology Criteria for Adverse Events (CTCAE) v5

Figure 1: Study Schema

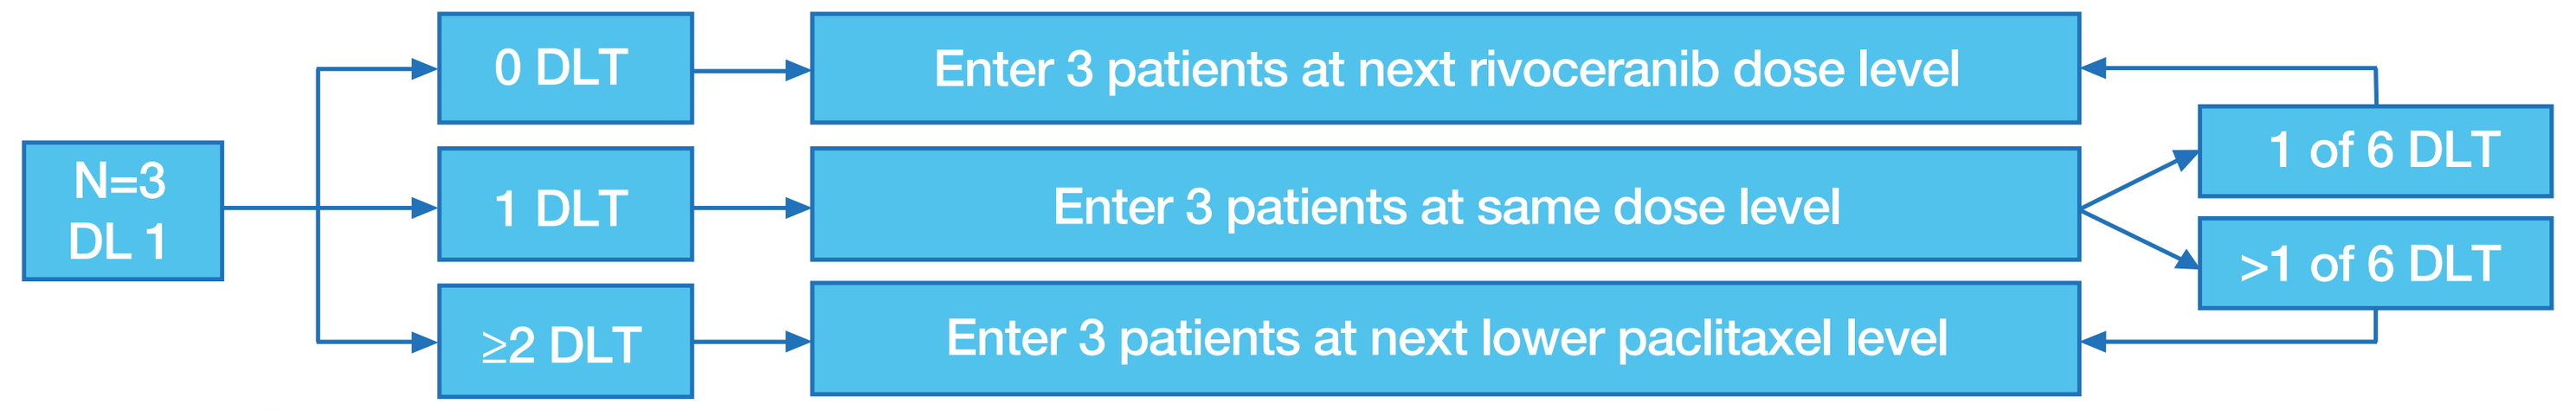
N=3
DL 1
0 DLT
1 DLT
≥2 DLT
Enter 3 patients at next rivoceranib dose level
Enter 3 patients at same dose level
Enter 3 patients at next lower paclitaxel level
1 of 6 DLT
>1 of 6 DLT

DL, dose level; DLT, dose limiting toxicity

Key Eligibility Criteria

- ≥19 years of age
- Locally advanced unresectable or metastatic gastric or GEJ cancer
- Disease refractory to or relapsing after first-line platinum and fluoropyrimidine-containing chemotherapy (with or without trastuzumab)
 - If disease progression occurred during or within 6 months after completion of adjuvant chemotherapy, that therapy was considered a first-line chemotherapy for eligibility
- Eastern Cooperative Oncology Group (ECOG) performance status 0 or 1
- No prior taxane
- No prior vascular endothelial growth factor (VEGF) pathway inhibitor
- No unstable central nervous system metastases
- No history of uncontrolled hypertension

Endpoints

- Primary phase 1 endpoints were the incidence of dose-limiting toxicity during cycle 1, adverse events (AEs), and serious adverse events (SAEs)
- Secondary phase 1 endpoints were pharmacokinetics, objective response rate (ORR), progression-free survival (PFS), overall survival (OS), disease control rate (DCR), and duration of response (DOR)

RESULTS

PATIENTS

- 12 patients were enrolled (**Table 1**)
 - Dose level 1 (rivoceranib 400 mg orally once daily + paclitaxel 80 mg/m² days 1, 8, and 15 every 28 days) initially enrolled 3 patients, and 1 patient experienced a DLT; therefore, another 3 additional patients were enrolled. No additional DLTs were observed
 - Dose level 2 (rivoceranib 500 mg orally once daily + paclitaxel 80 mg/m² days 1, 8, and 15 every 28 days) initially enrolled 3 patients, and 1 patient experienced a DLT; therefore, another 3 patients were enrolled. No additional DLTs were observed
 - The investigators and sponsor agreed not to escalate to dose level 3 because patients experienced DLTs at dose level 1 and dose level 2

Table 1: Baseline Characteristics

	Dose Level 1 (Rivoceranib 400 mg) n=6	Dose Level 2 (Rivoceranib 500 mg) n=6	All Patients N=12
Median age, year (range)	60.5 (37-67)	50.5 (36-65)	56 (36-67)
Male, n (%)	3 (50.0)	5 (83.3)	8 (66.7)
Asian, n (%)	6 (100)	6 (100)	12 (100)
ECOG PS 1, n (%)	6 (100)	6 (100)	12 (100)
Prior surgery, n (%)	4 (66.7)	5 (83.3)	9 (75.0)
Prior chemotherapy, n (%)	6 (100)	6 (100)	12 (100)
Oxaliplatin	4 (66.7)	6 (100)	10 (83.3)
Capecitabine	4 (66.7)	5 (83.3)	9 (75.0)
Cisplatin	2 (33.3)	0	2 (16.7)
Fluorouracil	1 (16.7)	1 (16.7)	2 (16.7)
Tegafur	1 (16.7)	0	1 (8.3)
Prior trastuzumab, n (%)	1 (16.7)	0	1 (8.3)

ECOG PS, Eastern Cooperative Oncology Group Performance Status.

SAFETY

- DLTs occurred at each dose level
 - Dose level 1: grade 3 oral mucositis (n=1)
 - Dose level 2: grade 3 febrile neutropenia (n=1); grade 3 gastric hemorrhage (n=1)
- All patients had at least one adverse event (AE) (**Table 2**)
 - 91.7% had at least one grade ≥3 AE
 - 16.7% had at lease one rivoceranib dose reduction for AEs
 - 41.7% had at least one paclitaxel dose reduction for AEs
- The most common grade ≥3 AEs were neutropenia (66.7%) and leukopenia (25.0%)
- Grade ≥3 AEs were more frequent in dose level 2

Table 2: Adverse Events

	Dose Level 1 (Rivoceranib 400 mg) n=6	Dose Level 2 (Rivoceranib 500 mg) n=6	All Patients N=12
Any AE	6 (100)	6 (100)	12 (100)
Grade ≥3 AE	5 (83.3)	6 (100)	11 (91.7)
AE leading to discontinuation of rivoceranib	0	0	0
Serious AE	2 (33.3)	1 (16.7)	3 (25.0)
Grade ≥3 AE occurring in >10% of patients overall			
Neutropenia	3 (50.0)	5 (83.3)	8 (66.7)
Leukopenia	1 (16.7)	2 (33.3)	3 (25.0)
Anemia	1 (16.7)	1 (16.7)	2 (16.7)
Peripheral neuropathy	1 (16.7)	1 (16.7)	2 (16.7)
Hypertension	1 (16.7)	1 (16.7)	2 (16.7)

AE, adverse event.

EFFICACY

- Of the 12 patients enrolled, 10 patients were measurable for response
- 6 patients had a partial response for an ORR of 60% (**Table 3**)
- The DCR was 100%

Table 3: Best Overall Responses (Response Evaluable Population)

	Dose Level 1 (Rivoceranib 400 mg) n=6	Dose Level 2 (Rivoceranib 500 mg) n=4	All Patients N=10
ORR, n (%)	4 (66.7)	2 (50)	6 (60)
PR	4 (66.7)	2 (50)	6 (60)
SD >12 weeks	2 (33.3)	2 (50)	4 (40)
DCR,[a] n (%)	6 (100)	4 (100)	10 (100)

DCR, disease control rate; ORR, objective response rate; PR, partial response; SD, stable disease
[a]DCR, CR+PR+SD for >12 weeks

- Median PFS was 8.5 months in the dose level 1 cohort and 6.0 months in the dose level 2 cohort (**Figure 2A**)
- Median OS was 16 months in the dose level 1 cohort and 8 months in the dose level 2 cohort (**Figure 2B**)

Figure 2. Kaplan-Meier Estimates of PFS (A) and OS (B)

A. Progression-Free Survival

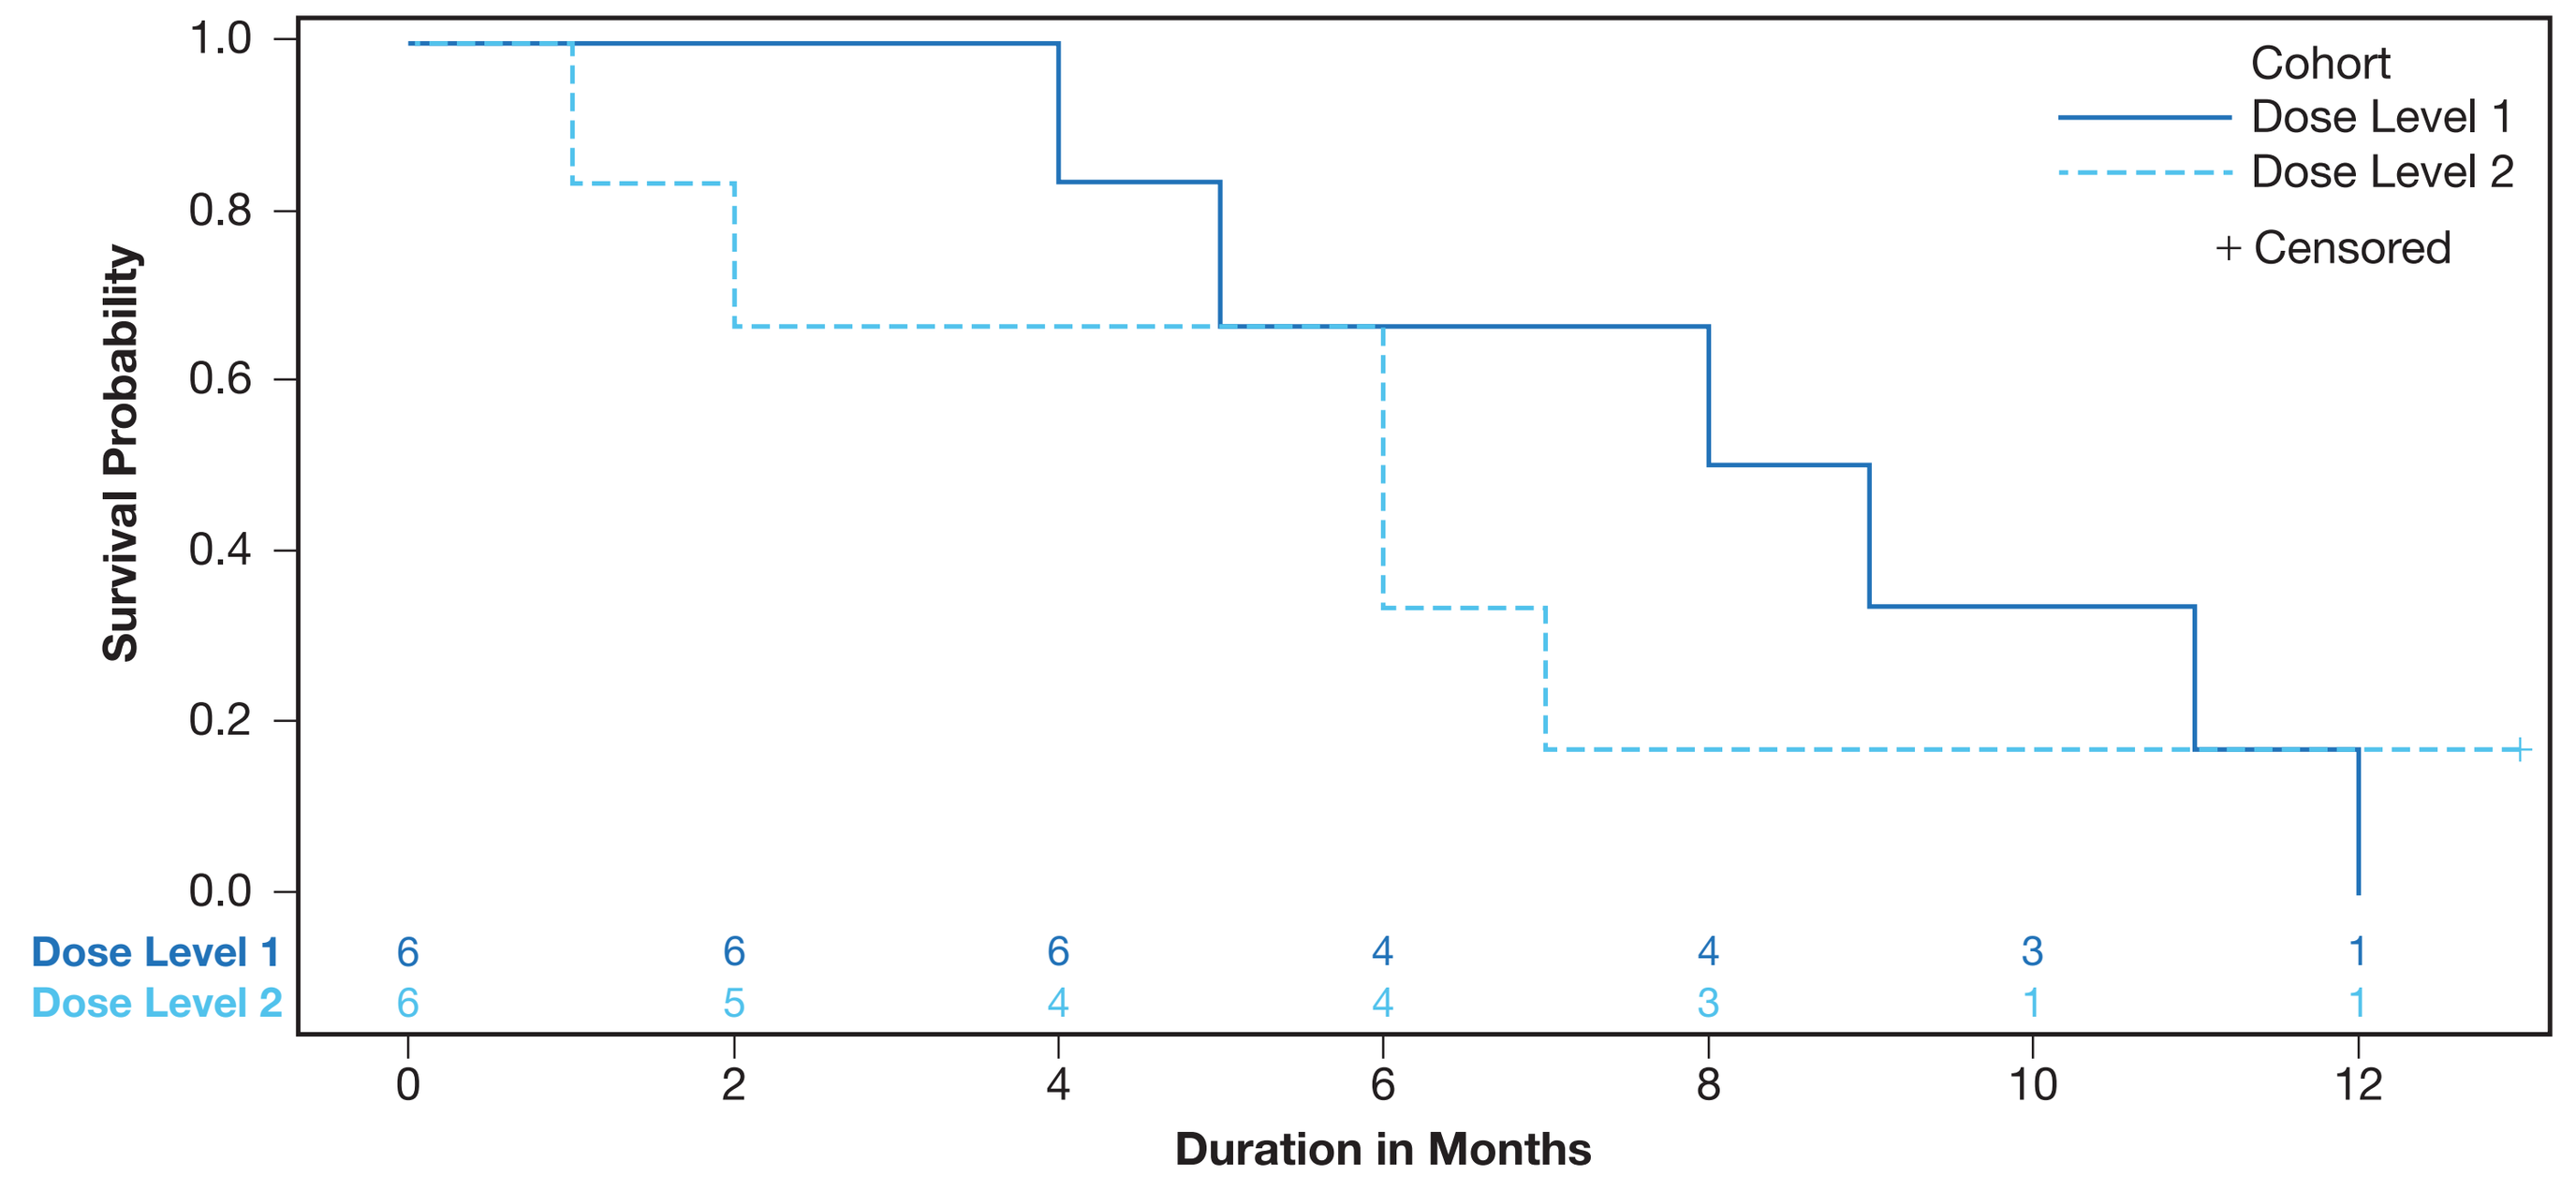
Survival Probability
1.0
0.8
0.6
0.4
0.2
0.0
Cohort
Dose Level 1
Dose Level 2
+ Censored
Dose Level 1 6 6 6 4 4 3 1
Dose Level 2 6 5 4 4 3 1 1
0 2 4 6 8 10 12
Duration in Months

B. Overall Survival

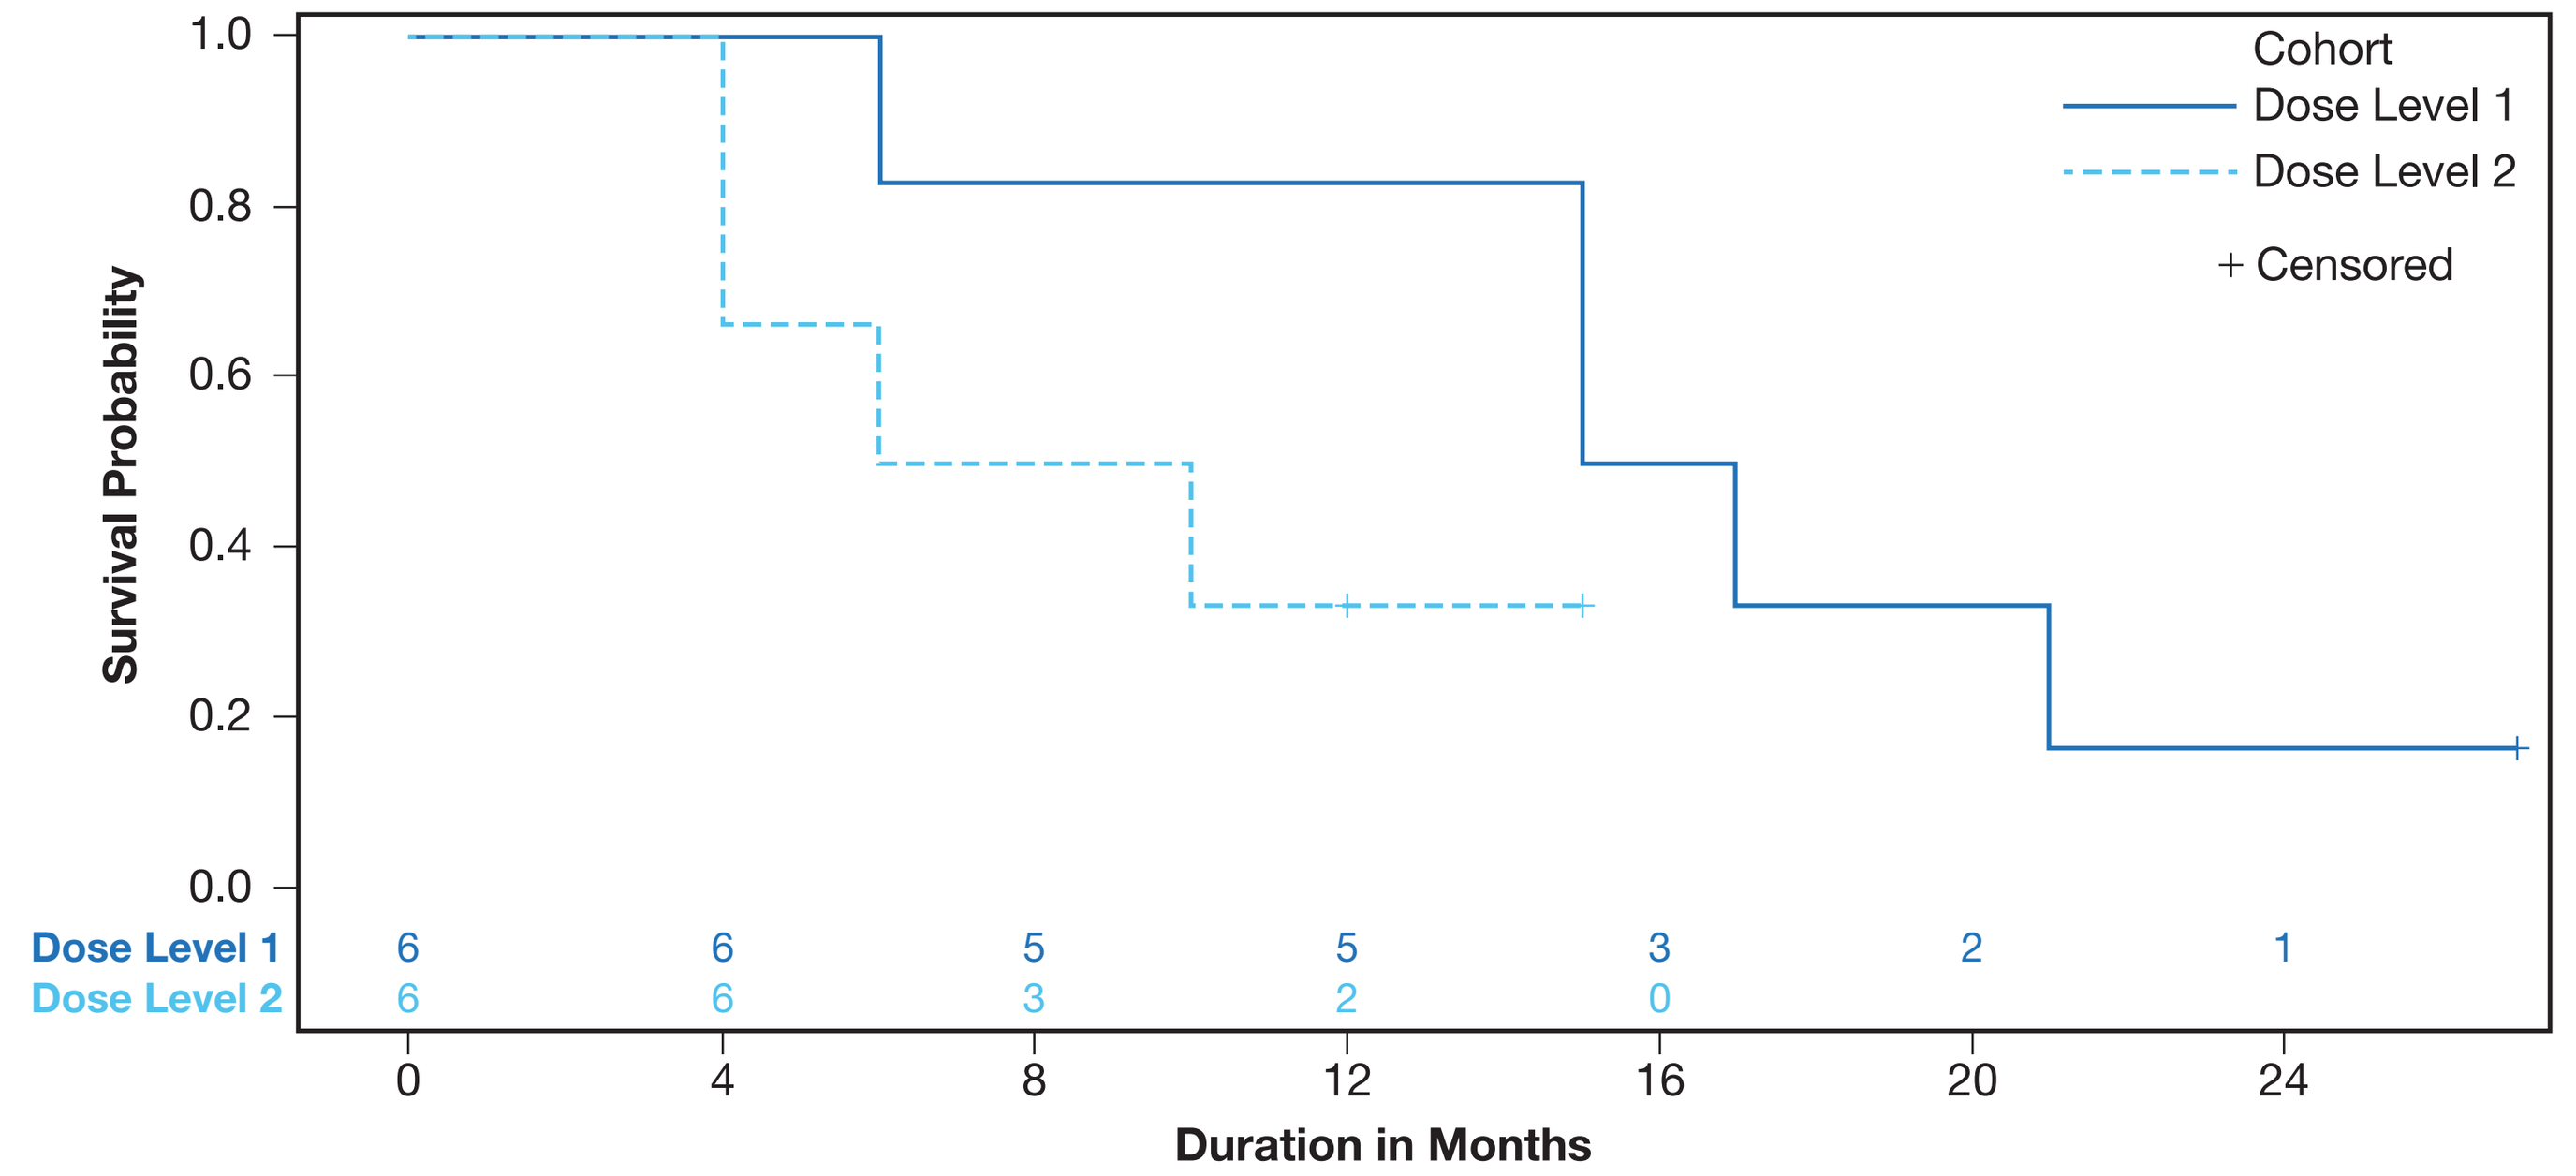
Survival Probability
1.0
0.8
0.6
0.4
0.2
0.0
Cohort
Dose Level 1
Dose Level 2
+ Censored
Dose Level 1 6 6 5 5 3 2 1
Dose Level 2 6 6 3 2 0
0 4 8 12 16 20 24
Duration in Months

PHARMACOKINETICS

- Cross-study comparison with the PK from paclitaxel monotheray suggests no apparent drug-drug interaction with rivoceranib;[1,6,7] however, further investigation is needed before a definitive conclusion regarding drug-drug interactions can be made

Figure 3. Mean Plasma Drug Concentrations, Linear Scale

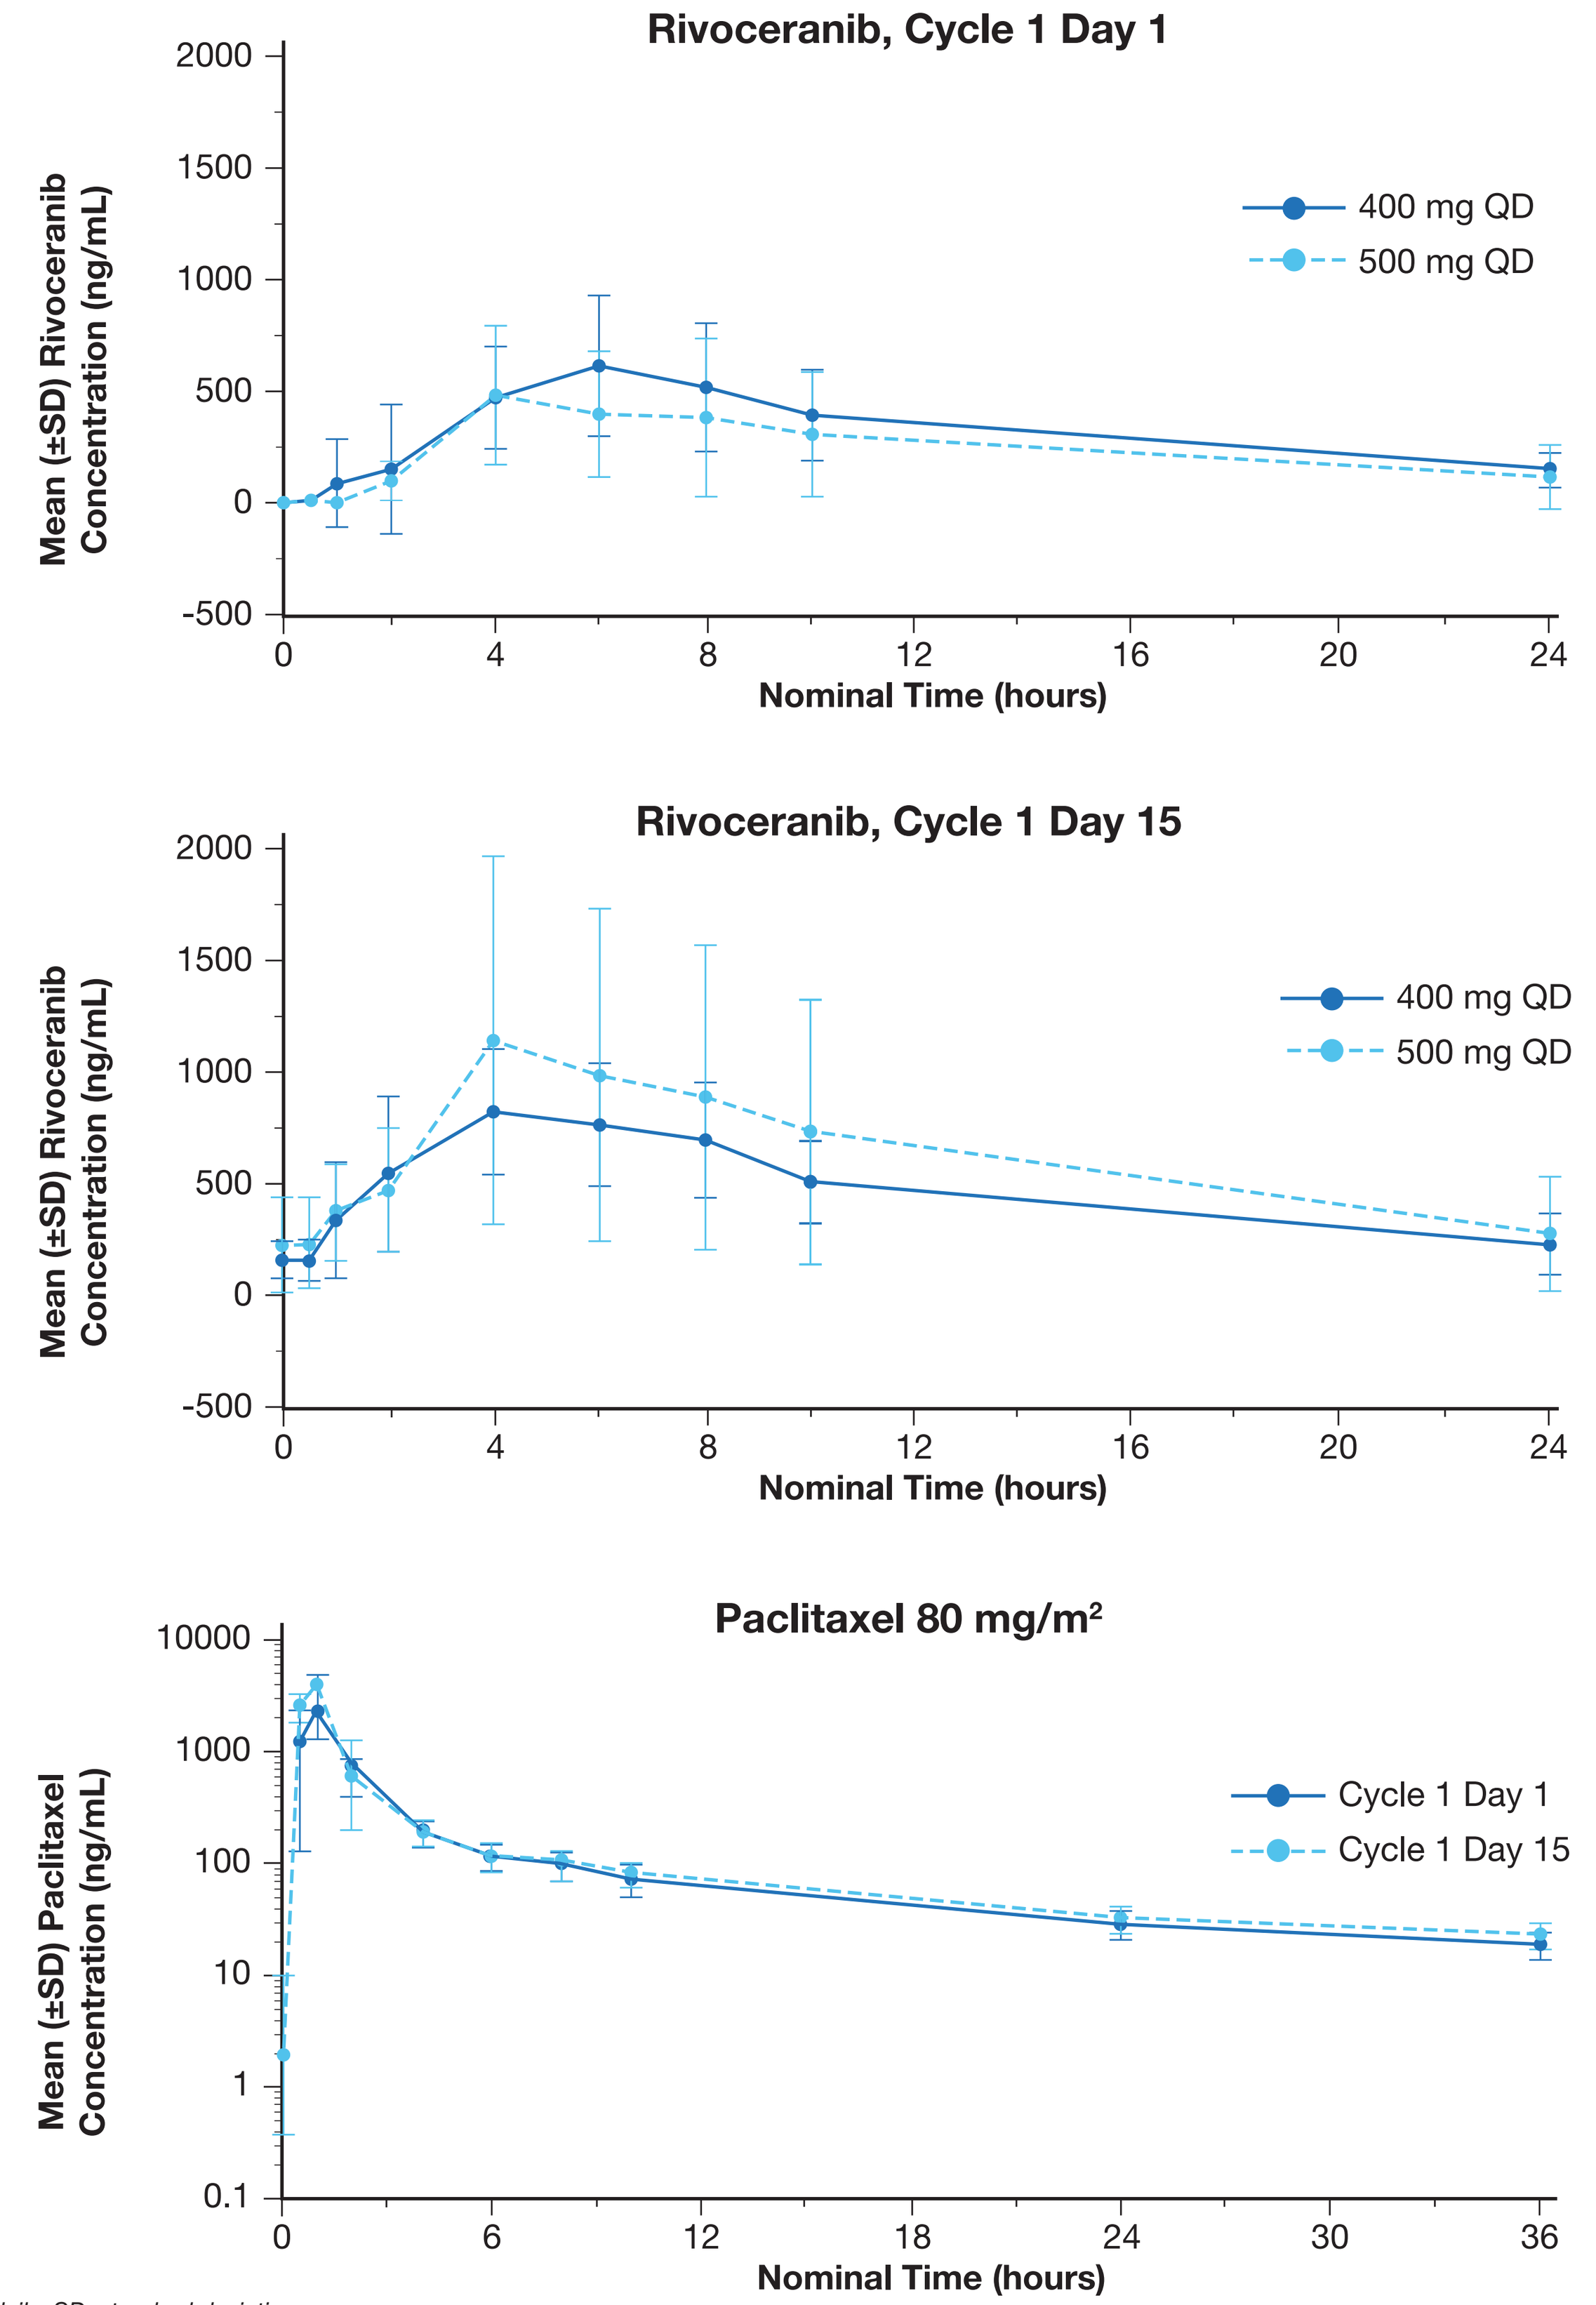
Rivoceranib, Cycle 1 Day 1
Mean (±SD) Rivoceranib Concentration (ng/mL)
2000
1500
1000
500
0
-500
400 mg QD
500 mg QD
0 4 8 12 16 20 24
Nominal Time (hours)
Rivoceranib, Cycle 1 Day 15
Mean (±SD) Rivoceranib Concentration (ng/mL)
2000
1500
1000
500
0
-500
400 mg QD
500 mg QD
0 4 8 12 16 20 24
Nominal Time (hours)
Paclitaxel 80 mg/m²
Mean (±SD) Paclitaxel Concentration (ng/mL)
10000
1000
100
10
1
0.1
Cycle 1 Day 1
Cycle 1 Day 15
0 6 12 18 24 30 36
Nominal Time (hours)

QD, once daily; SD, standard deviation.

CONCLUSIONS

- The combination of rivoceranib + paclitaxel as 2nd-line treatment in patients with advanced gastric or GEJ cancer showed good clinical activity with a manageable safety profile
- Considering the safety profile and efficacy trend, the recommended phase 2 dose was defined as rivoceranib 400 mg + paclitaxel 80 mg/m²
- Pharmacokinetic data suggest a lack of drug-drug interaction between rivoceranib and paclitaxel
- Based on these data, the combination warrants further investigation

REFERENCES: 1. Sachar M, et al. *Fundam Clin Pharmacol*. 2021;35:485–495; **2.** Sachar M, et al. *Fundam Clin Pharmacol*. 2022;36:171–181; **3.** Shaik F, et al. *Biomolecules.* 2020;10:1673; **4.** Tian Z, et al. *Front Oncol*. 2021;11:749083; **5.** Xu Z, et al. *Cancer Manag Res*. 2019;11:4905-4915; **6.** Ding J, et al. *Drug Metab Dispos*. 2013;41:1195-1210; **7.** Stage TB, et al. *Clin Pharmacokinet*. 2018;57:7-19.

ACKNOWLEDGEMENTS: Editorial assistance was provided by Phillips Gilmore Oncology Communications, Inc. and funded by Elevar Therapeutics.

CONTACT FOR POSTER INFORMATION/QUESTIONS: medicalinformation@elevartherapeutics.com. **Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.**